Analysis of percent drug dissolve with dissolution tester

Suchanya Udomrattanayothin[1], Kongkiat Trisuwan[1], Warittha Kuntiang[2]

[1]Department of Chemistry, Faculty of Science, Chiang Mai University, Chiang Mai 50200, Thailand
[2]Siam Bheasach Co., Ltd. 123 Soi Chokchai Ruammit, Rd. Vibhavadi Rangsit, Chomphon Suc-district, Chatuchak District, Bangkok 10900, Thailand

ABSTRACT

One of the most important factors in the effectiveness of oral drugs is their dissolution after they enter the body. From studying the working principles and determining the dissolution values of 6 tablets each of capsules X, tablets Y, and suspensions Z using a Dissolution tester that simulates the dissolution conditions of drugs in the stomach. Therefore, the medium which is part of the gastric pH was selected to determine the dissolution value of drug, which results were in accordance with the criteria and principles of the instruments specified by the international Pharmacopoeia.

INTRODUCTION

Oral drugs of the Immediate release (IR) type, which require drug dissolution testing, must pass through a random sampling process to test the quality according to the criteria and principles of the equipment specified in the international Pharmacopoeia, using a Dissolution tester that simulates the dissolution conditions of drugs in the stomach. and interpreted using UV and HPLC techniques.

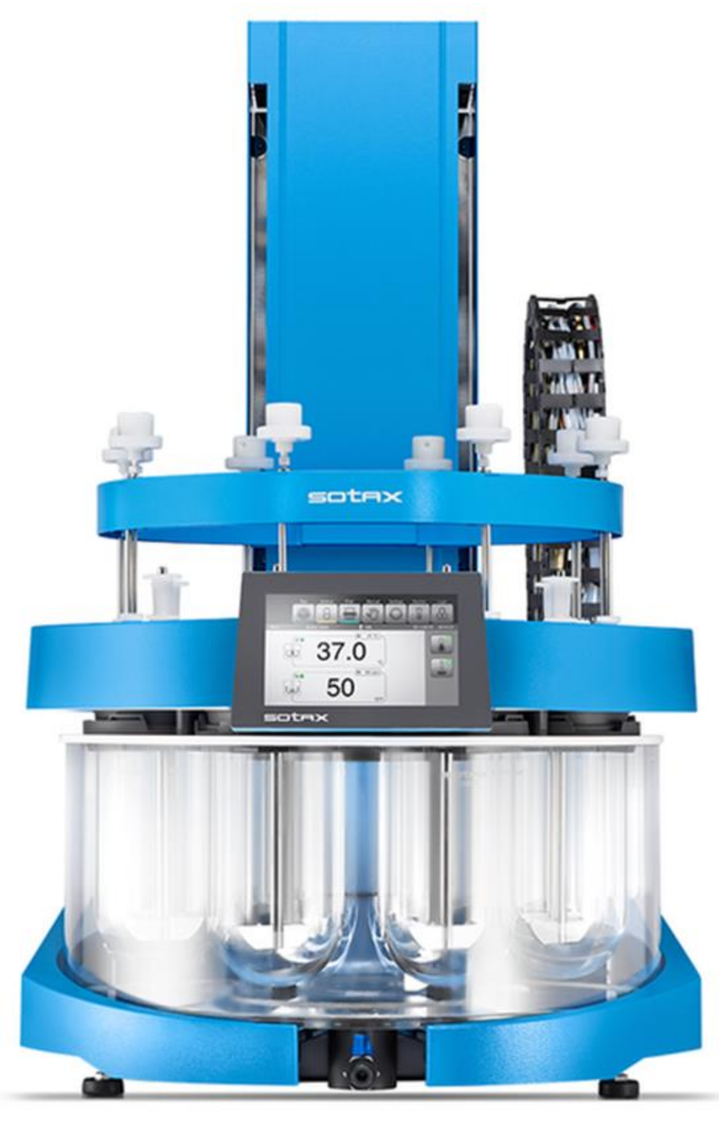

METHODOLOGY

Capsule X

Medium

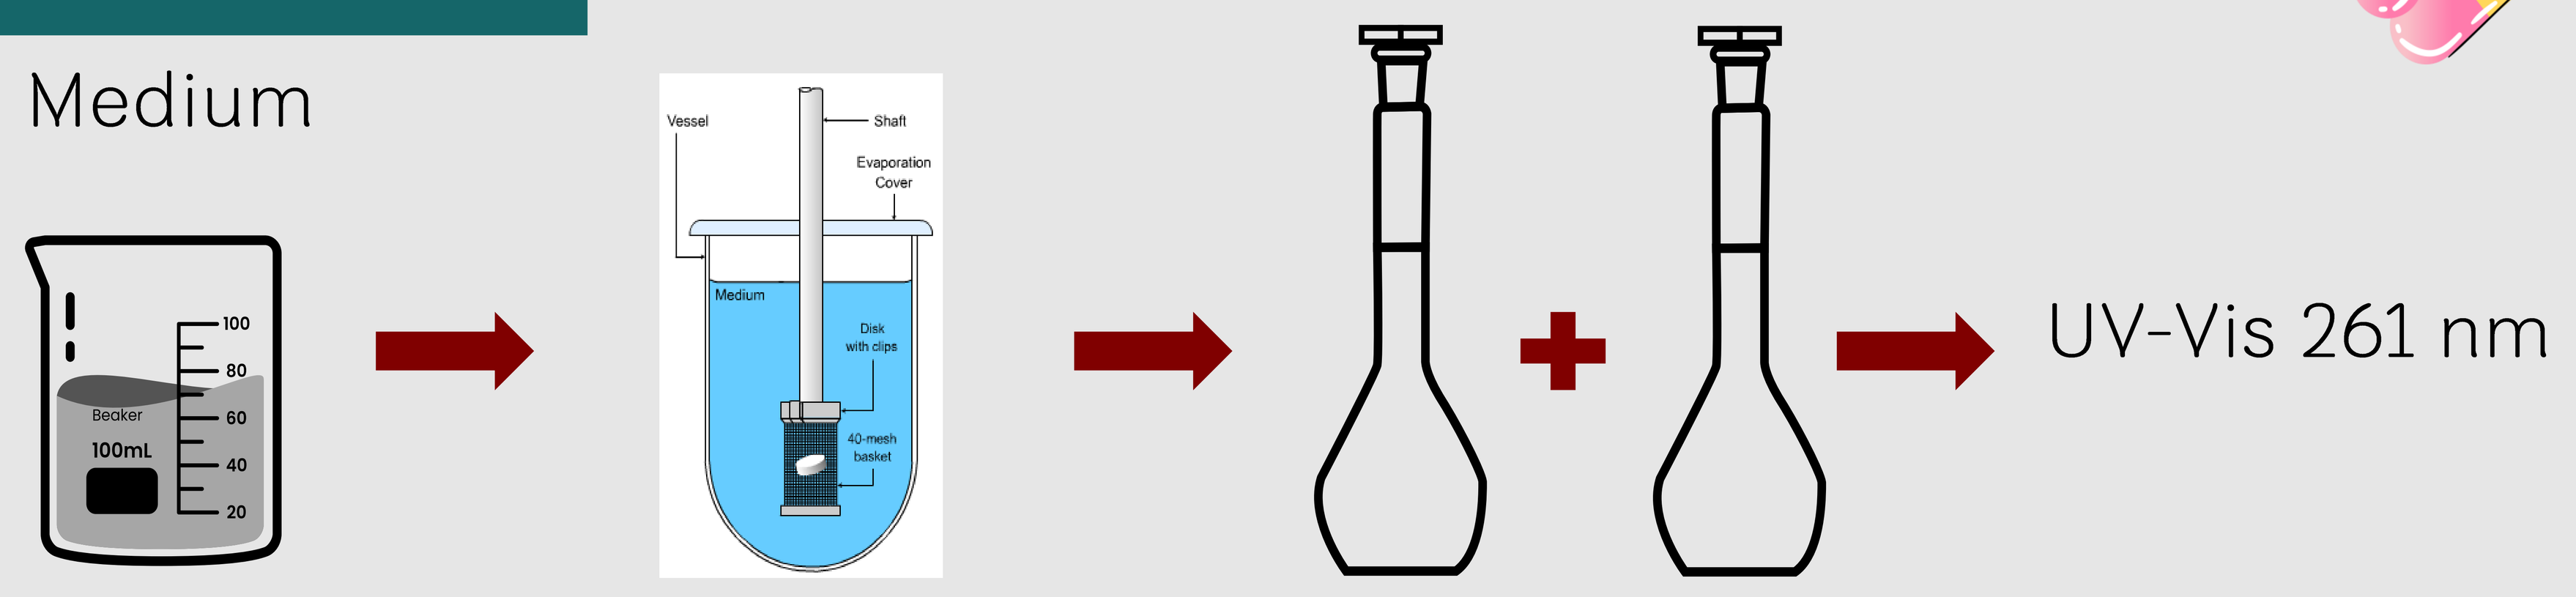
UV-Vis 261 nm

0.1 N HCl

Apparatus I
Speed : 100 rpm
Time : 45 min

Sample STD

Tablet Y

Medium

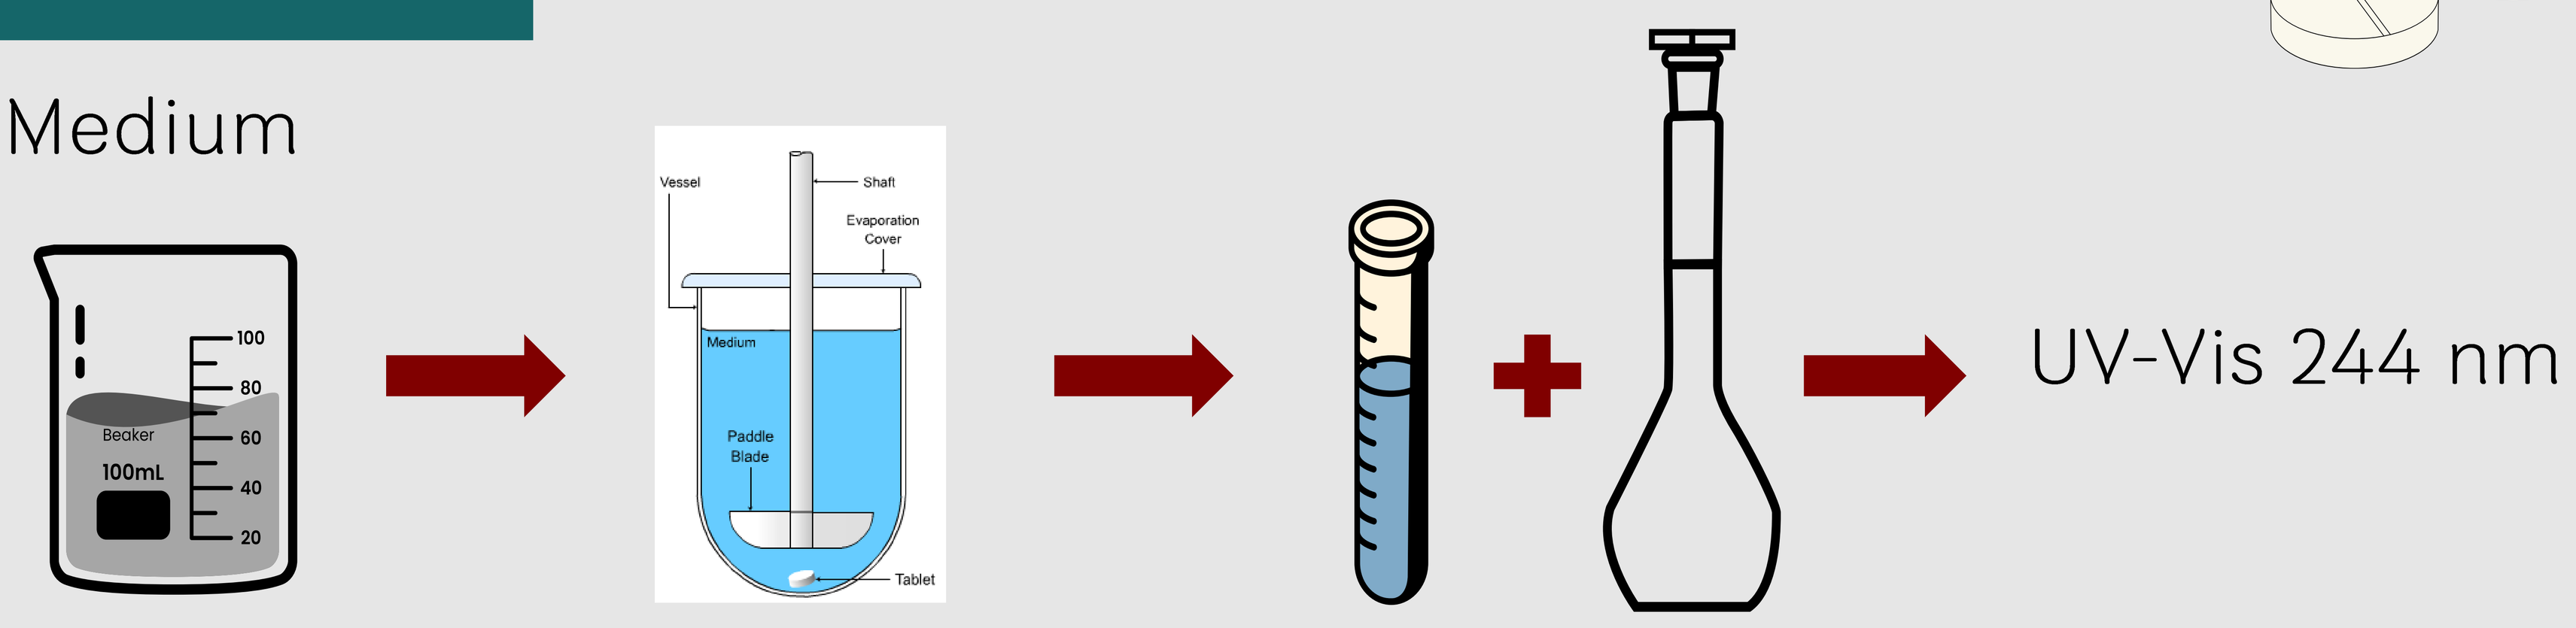
UV-Vis 244 nm

Phosphate buffer pH 6.8

Apparatus II
Speed : 75 rpm
Time : 15 min

Sample STD

Suspension Z

Medium

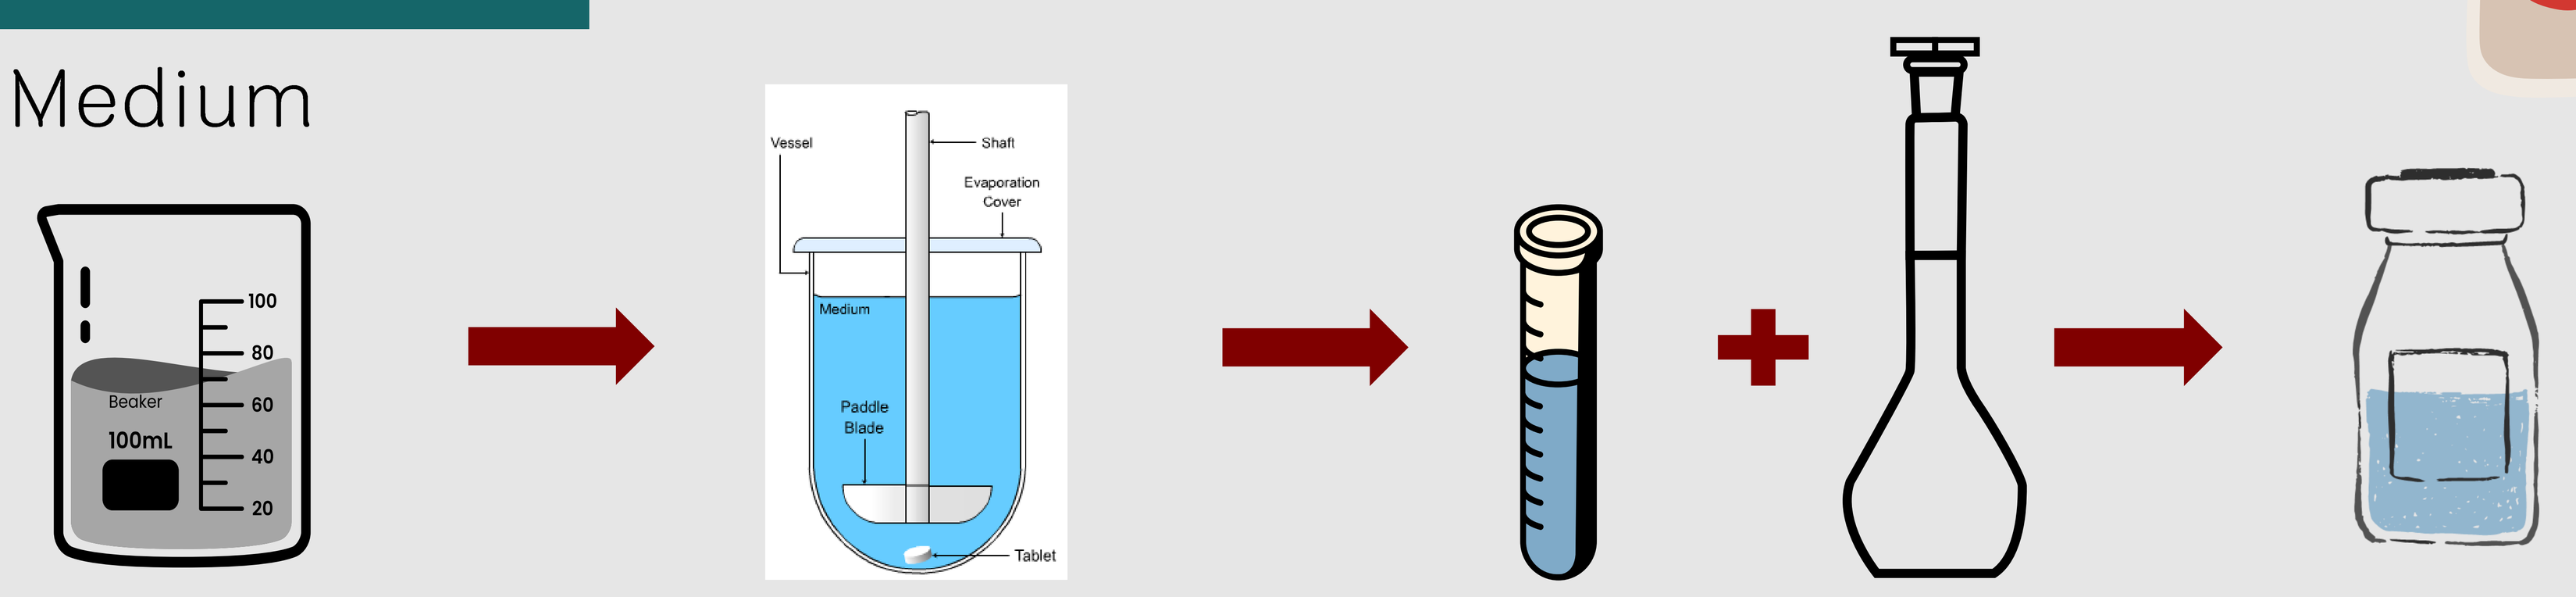

Sodium phosphate buffer pH 6.0

Apparatus II
Speed: 50 rpm
Time: 30 min

Sample STD HPLC

ACKNOWLEDGMENT

The authors would like to offer the special thanks to Assoc. Prof. Dr. Kongkiat Trisuwan (Advisor) for their professional guidance and valuable support. In addition, the authors would like to extend thanks to Siam Bheasach Co., Ltd. for providing the equipments for material characterzation and testing

ACCEPTANCE CRITERIA

Stage	Number of test	Acceptance Criteria
S_1	6	Each unit is not less than Q + 5%
S_2	6	Average of 12 units (S_1+S_2) is equal to or greater than Q, and no unit is less than Q – 15%

The Q value is the solubility value of a drug specified in pharmacopoeia standards.

RESULTS AND DISCUSSION

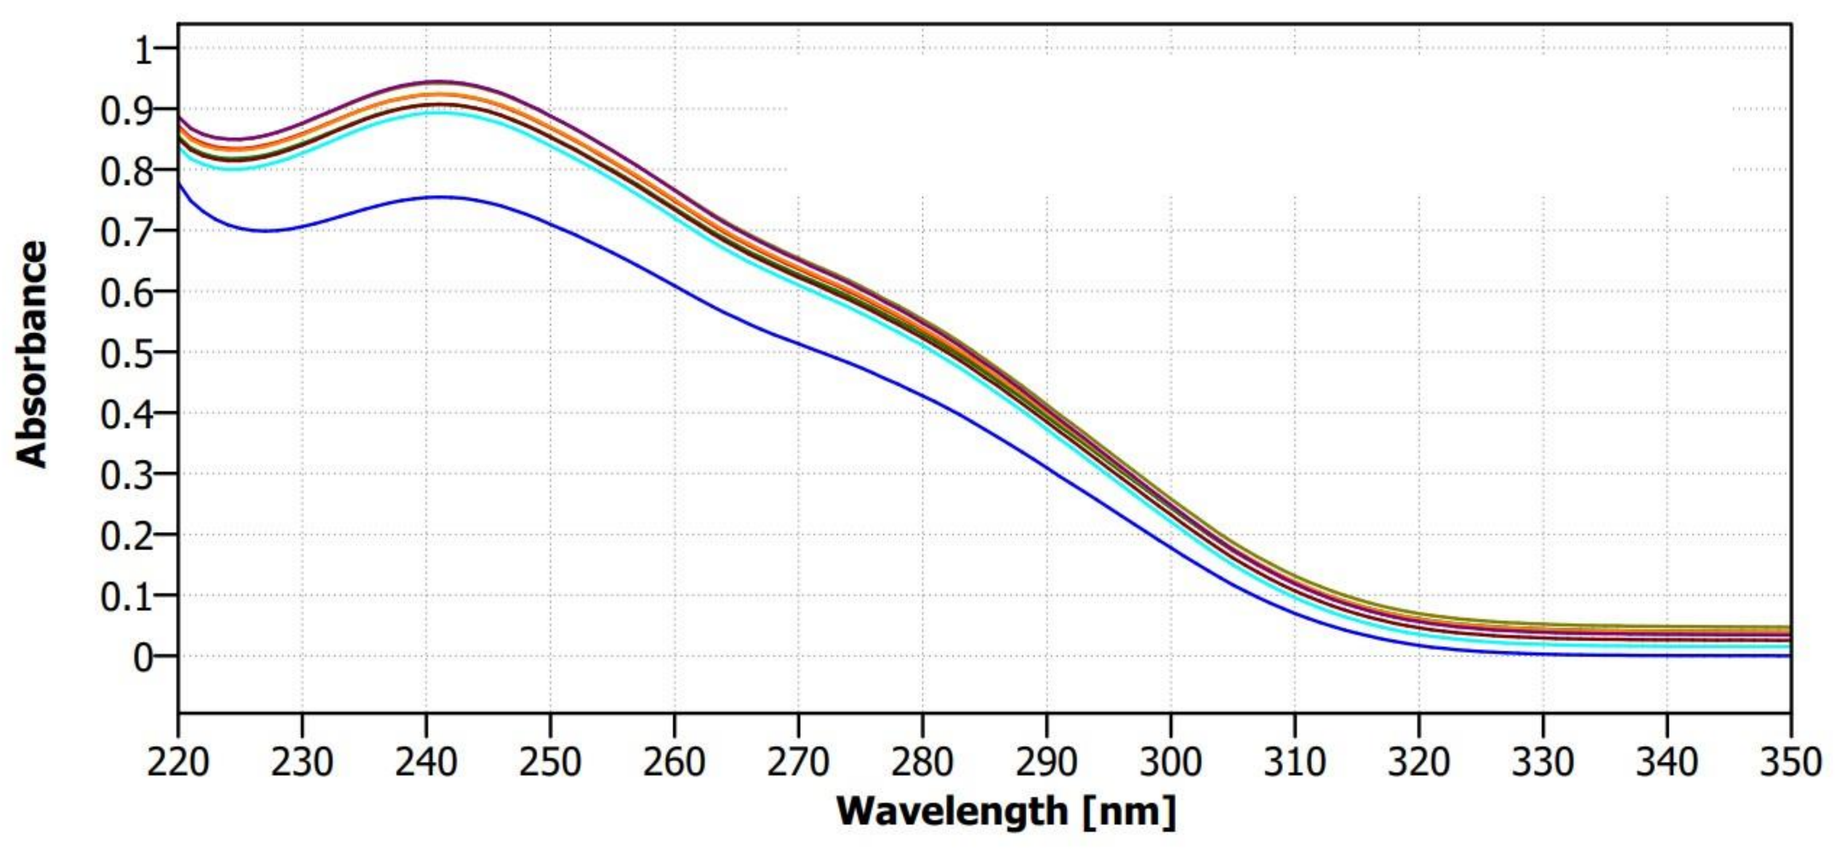

Fig.1 Absorbance of Tablet Y at UV-Vis 261 nm

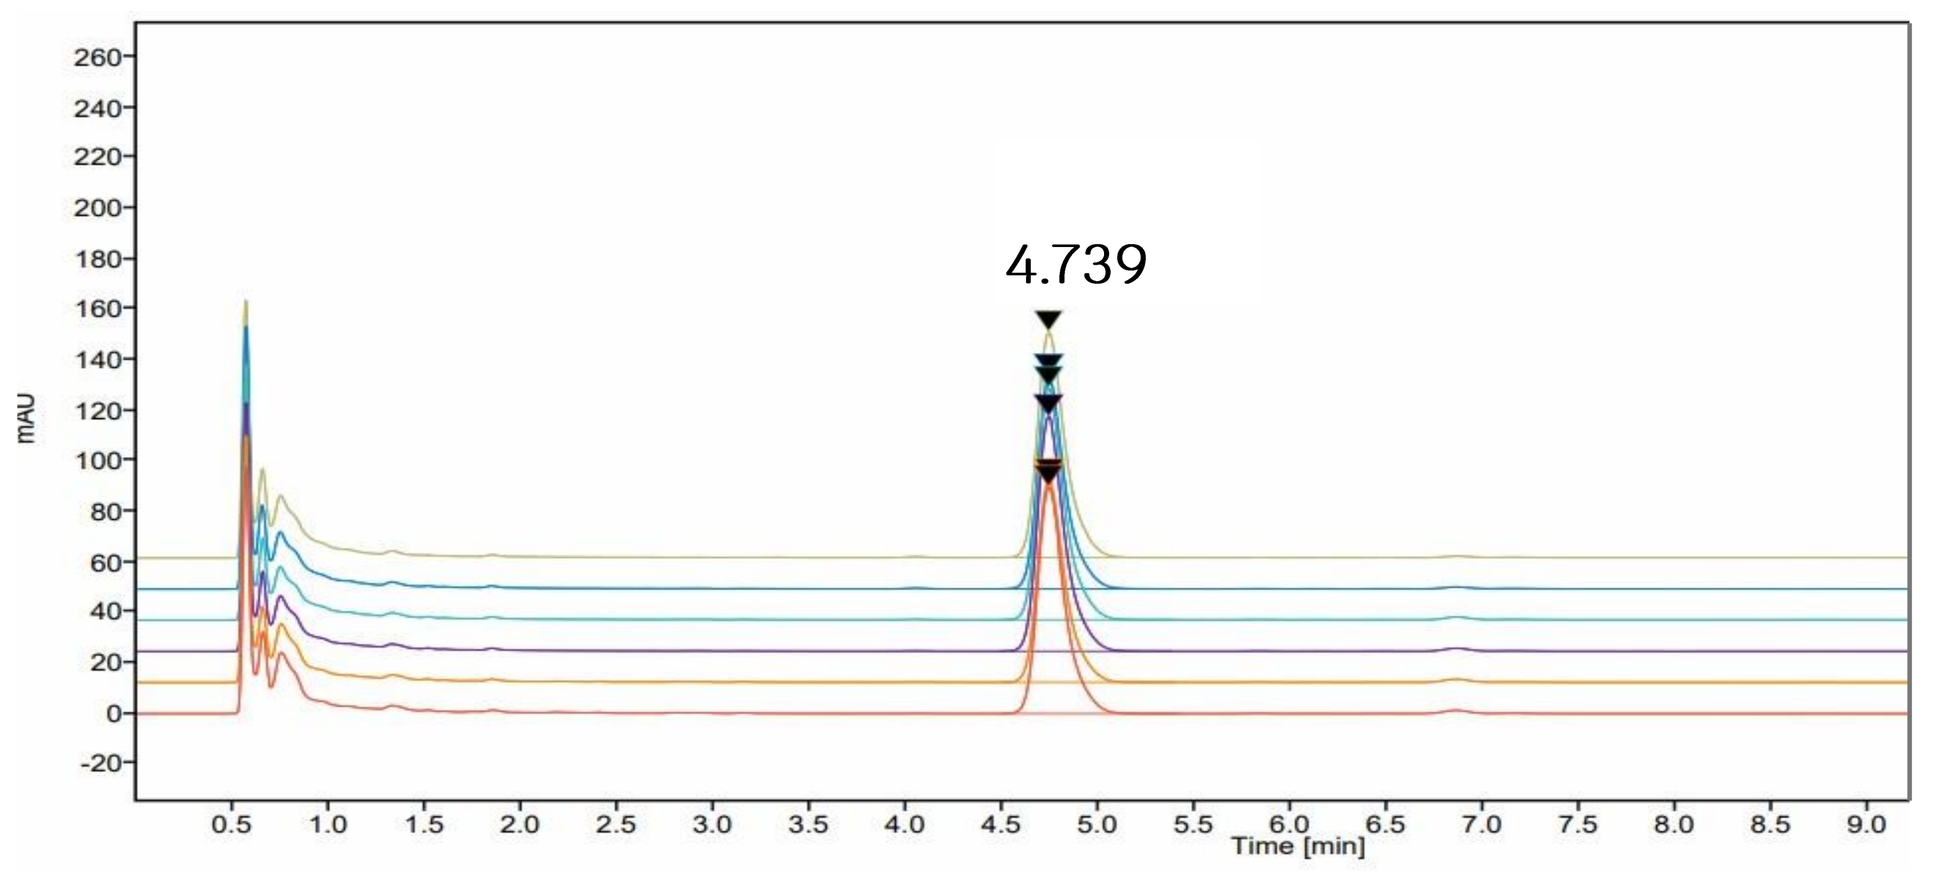
4.739

Fig.2 Absorbance of Suspension Z of HPLC

Table of % Dissolution of drug at various time

Sample	Capsule X 45 min	Tablet Y 15 min	Suspension Z 30 min
1	95.50	95.12	95.89
2	93.91	91.15	90.24
3	95.33	95.03	96.97
4	95.36	96.53	97.90
5	93.62	94.62	85.16
6	95.11	95.90	94.99

The Q value of Capsule X is 75, Tablet Y is 80 and Suspension Z is 75

CONCLUSION

From the dissolution test of the drug, which is an important test for controlling the quality of oral drugs, a study was conducted on the working principle and use of the Dissolution tester. Different instruments, preparations, interpretations, and analytical conditions were used. The experimental results were in accordance with the criteria and principles set by the pharmaceutical textbooks, and the tested samples had good dissolution capability.

REFERENCE

[1] Pharm. Patima Maneesathit. Control of drug dissolution in oral tablets (Department of Medical Sciences, Ministry of Public Health). Retrieved from https://ccpe.pharmacycouncil.org.
[2] Honorary Prof. Dr. Pharm Somphon Prakongphan, Dissolution test of oral solids (Royal Academy, Bureau of Science, Royal Society). Retrieved from https://ccpe.pharmacycouncil.org.